1893

Efficacy of isatuximab with pomalidomide and dexamethasone in elderly patients with relapsed/refractory multiple myeloma: ICARIA-MM subgroup analysis

Fredrik H. Schjesvold,[1] Paul G. Richardson,[2] Michel Attal,[3] Thierry Facon,[4] Adrián Alegre,[5] Andrew Spencer,[6] Artur Jurczyszyn,[7] Kazutaka Sunami,[8] Laurent Frenzel,[9] Chang-Ki Min,[10] Solenn Le-Guennec,[11] Frank Campana,[12] Helgi van de Velde,[12] Samira Bensfia,[13] Sara Bringhen[14]

[1]Oslo Myeloma Center, Oslo University Hospital and KG Jebsen Center for B cell malignancies, University of Oslo, Oslo, Norway; [2]Dana-Farber Cancer Institute, Boston, MA; [3]Institut Universitaire du Cancer de Toulouse-Oncopole, Toulouse, France; [4]Lille University Hospital, Lille, France; [5]Hospital Universitario La Princesa, Madrid, Spain; [6]Department of Clinical Haematology, Alfred Health-Monash University, Melbourne, Australia; [7]Department of Hematology, Jagiellonian University Medical College, Cracow, Poland; [8]Department of Hematology, National Hospital Organization Okayama Medical Center, Okayama, Japan; [9]Hopital Necker-Enfants Malades, Paris, France; [10]Department of Hematology, Catholic Hematology Hospital and Leukemia Research Institute, Seoul St. Mary's Hospital, College of Medicine, The Catholic University of Korea, Seoul, Korea, Republic of (South); [11]Sanofi R&D, Vitry-sur-Seine, France; [12]Sanofi R&D, Cambridge, MA; [13]Sanofi Global Oncology, Cambridge, MA; [14]Myeloma Unit, Division of Hematology, University of Torino, Azienda-Ospedaliero Universitaria Città della Salute e della Scienza di Torino, Torino, Italy

BACKGROUND

- Multiple myeloma (MM) is most frequently diagnosed in people aged 65–74 years with a median age at diagnosis of 69 years.[1]
- The treatment of elderly patients is challenging primarily due to frailty, comorbidities, and decreased resilience to treatment-related toxicity.[2] These patients are frequently underrepresented in clinical trials as they often fail to meet the strict eligibility criteria.[2]
- Advanced age negatively impacts the prognosis of patients with MM.[3]
- Isatuximab (Isa), an IgG1 monoclonal antibody that targets a specific epitope of CD38, has multiple mechanisms of action for killing tumor cells,[4,5] and its activity has been shown to be augmented by pomalidomide.[6]
- ICARIA-MM (NCT02990338) was a randomized, open-label, active-controlled, multicenter phase 3 study that compared treatment of Isa in combination with pomalidomide (P) and dexamethasone (d) with Pd.[6]
 - Eligible patients had relapsed/refractory MM (RRMM) and ≥2 prior lines of therapy, including lenalidomide and a proteasome inhibitor.[6]
- In ICARIA-MM, median progression-free survival (PFS) was significantly improved with Isa-Pd vs Pd (11.53 vs 6.47 months) with a hazard ratio (HR) of 0.596 (95% confidence interval [CI] 0.436–0.814).[7]
- This prespecified subgroup analysis of ICARIA-MM compared the efficacy and safety of treatment in elderly patients (aged ≥75 years) with younger patients.

METHODS

- Patients were randomized 1:1 to Isa-Pd or Pd stratified by age (<75 vs ≥75 years) and number of prior lines of therapy (2–3 vs >3). The study design, including dosing information, is given in **Figure 1**.
- Prespecified analyses were performed according to three age groups: <65, 65–74 and ≥75 years.
- The cut-off date for PFS and OS analyses was October 11, 2018.

Study endpoints

- The primary endpoint was PFS, assessed by an independent review committee.
- Key secondary endpoints were overall response rate (ORR) and overall survival (OS).
- Treatment-emergent adverse events (TEAEs) were graded per National Cancer Institute Common Terminology Criteria for Adverse Events (NCI-CTCAE) v4.03.

Figure 1. ICARIA-MM study design

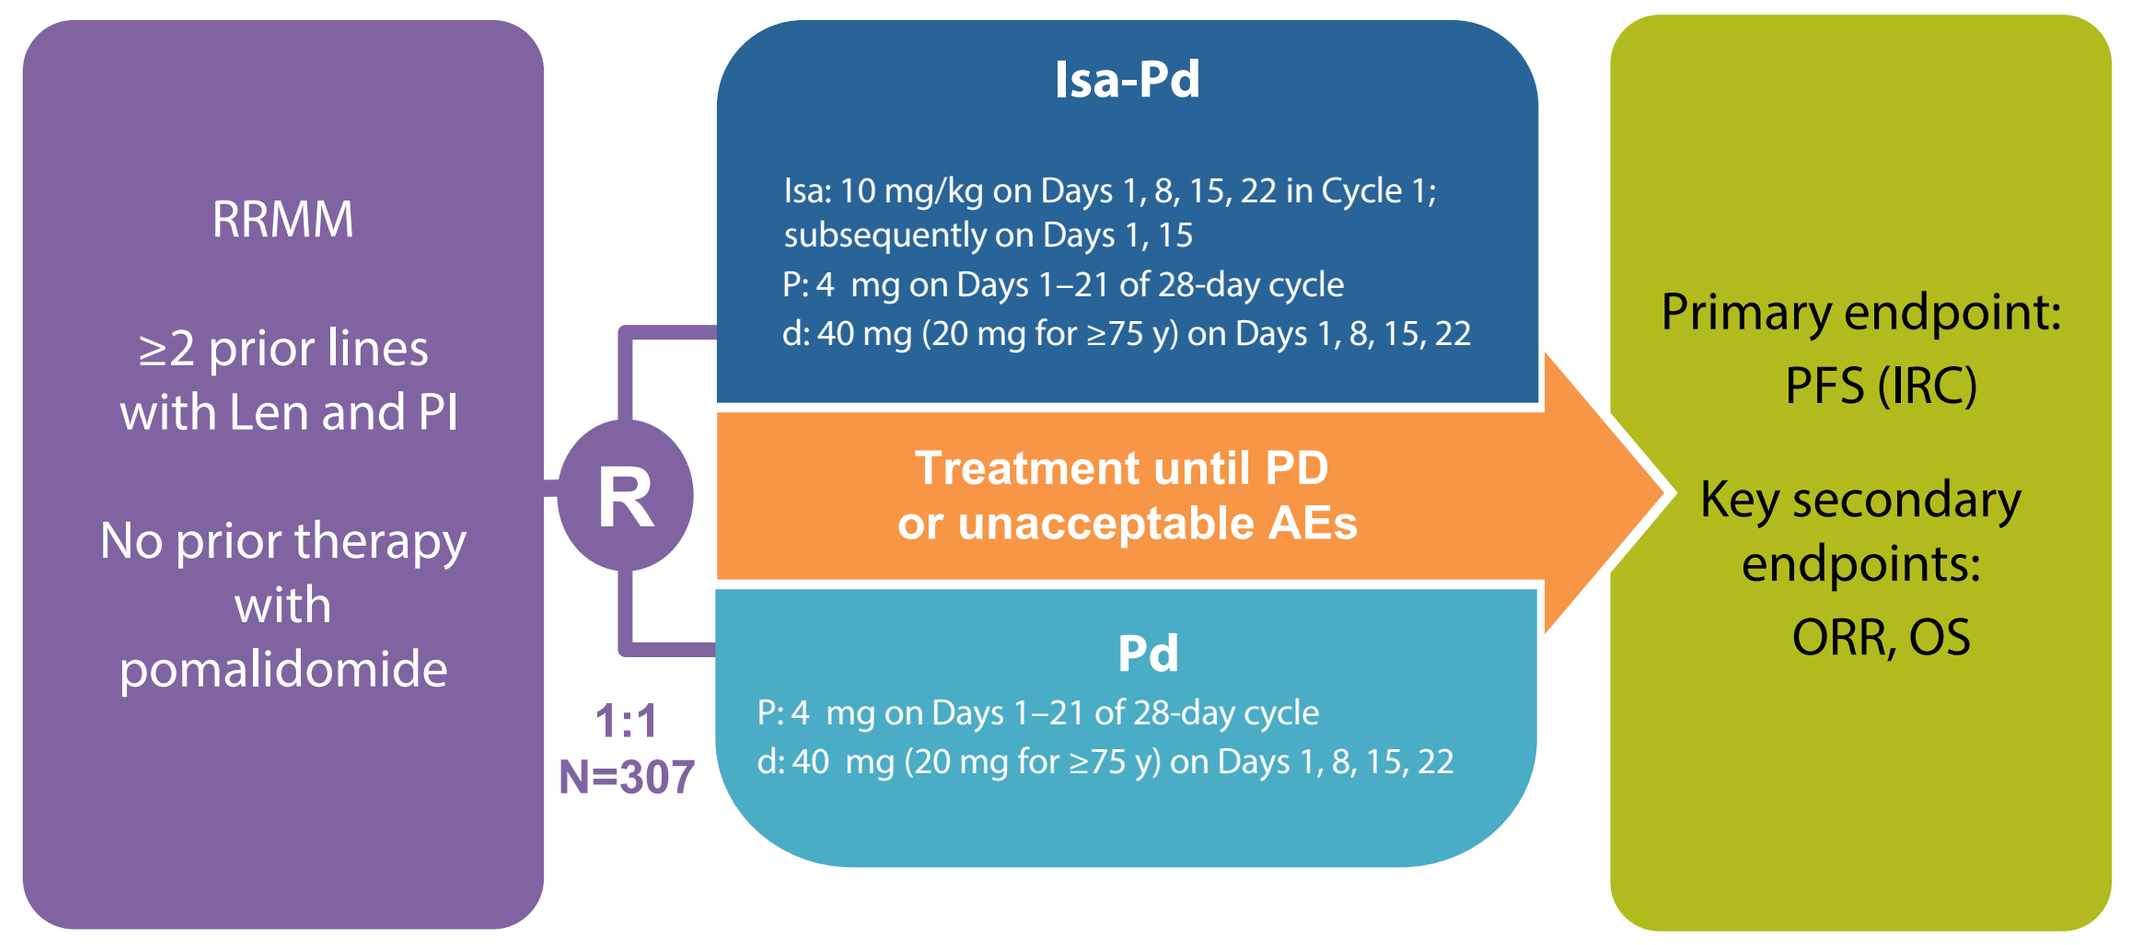

RRMM
≥2 prior lines with Len and PI
No prior therapy with pomalidomide
R
1:1
N=307
Isa-Pd
Isa: 10 mg/kg on Days 1, 8, 15, 22 in Cycle 1; subsequently on Days 1, 15
P: 4 mg on Days 1–21 of 28-day cycle
d: 40 mg (20 mg for ≥75 y) on Days 1, 8, 15, 22
Treatment until PD or unacceptable AEs
Pd
P: 4 mg on Days 1–21 of 28-day cycle
d: 40 mg (20 mg for ≥75 y) on Days 1, 8, 15, 22
Primary endpoint: PFS (IRC)
Key secondary endpoints: ORR, OS

Patients in the Isa-Pd arm received premedication before infusion that consisted of ranitidine (50 mg or equivalent), diphenhydramine (25 to 50 mg or equivalent), and acetaminophen (650 to 1000 mg). Dexamethasone was used both as part of premedication and as part of the backbone combination in the Isa-Pd arm.

AE, adverse event; d, dexamethasone; IRC, independent review committee; Isa, isatuximab; Len, lenalidomide; ORR, overall response rate; OS, overall survival; P, pomalidomide; PD, progressive disease; PFS, progression-free survival; PI, proteasome inhibitor; R, randomization; RRMM, relapsed/refractory multiple myeloma; y, years.

RESULTS

Patient baseline characteristics and disposition

- 307 patients were randomized to Isa-Pd (n=154) and Pd (n=153).
- Patient baseline characteristics have been presented previously.[7]
 - The median age of patients was 68.0 and 66.0 years in the Isa-Pd and Pd arms, respectively and the median number of prior lines was 3 for both treatment arms.
 - The proportion of elderly patients (aged ≥75 years) was similar between treatment arms (20.8% for Isa-Pd vs 19.0% for Pd).
 - The proportion of patients aged <65 years was lower in the Isa-Pd arm than the Pd arm (35.1% vs 45.8%) and the proportion of patients aged 65–74 years was higher in the Isa-Pd arm than the Pd arm (44.2% vs 35.3%).

Efficacy – PFS

- In the overall population, a significant benefit in PFS was observed with Isa-Pd compared with Pd. Median PFS was 11.53 and 6.47 months, respectively (HR 0.596; 95% CI 0.436–0.814; p=0.001).
- This significant benefit in PFS with Isa-Pd was also observed and was almost identical in all three patient age subgroups.
 - In patients aged ≥75 years, median PFS was 11.40 months (Isa-Pd; n=32) vs 4.47 months (Pd; n=29), HR 0.479; 95% CI 0.242–0.946 (**Figure 2**).
 - In patients aged 65–74 years, median PFS was 11.57 months (Isa-Pd; n=68) vs 8.58 months (Pd; n=54), HR 0.638; 95% CI 0.385–1.059 (**Figure 3**).
 - In patients aged <65 years, median PFS was 11.53 (Isa-Pd; n=54) vs 5.03 months (Pd; n=70), HR 0.656; 95% CI 0.401–1.074 **(Figure 4).**

Efficacy – ORR

- In the overall population, the ORR was 60.4% with Isa-Pd and 35.3% with Pd, with an odds ratio (OR) of 2.80 (95% CI 1.72–4.56; p<0.0001).
- **Figure 5** shows response rates by age group.
 - For patients aged ≥75 years, the ORR was 53.1% and 31.0% in the Isa-Pd and Pd arms, respectively (OR 2.52; 95% CI 0.79–8.26).
 - For patients aged 65–74 years, the ORR was 64.7% and 38.9% in the Isa-Pd and Pd arms, respectively (OR 2.88; 95% CI 1.29–6.46).
 - For patients aged <65 years, the ORR was 59.3% and 34.3% in the Isa-Pd and Pd arms, respectively (OR 2.79; 95% CI 1.26–6.20).

Efficacy – very good partial response (VGPR) and minimal residual disease (MRD) negativity

- Overall, 31.8% and 8.5% of patients achieved at least a VGPR (≥VGPR) with Isa-Pd and Pd, respectively (OR 5.03; 95% CI 2.51–10.59).
- Across age groups, the proportion of patients who achieved ≥VGPR was consistently higher with Isa-Pd vs Pd (**Figure 5**).
 - For patients aged ≥75 years, 31.3% of patients achieved ≥VGPR with Isa-Pd, while no patient achieved this with Pd (OR not calculable).
 - Corresponding ORs were 3.21 (95% CI 1.17–9.70) for patients aged 65–74 years and 4.90 (95% CI 1.64–16.35) for patients aged <65 years.
- Overall, eight patients in the Isa-Pd arm had MRD negativity (at 10^{-5}). Two of these patients were ≥75 years of age and two were aged 65–74 years. The remaining four patients were aged <65 years. No patients in the Pd arm achieved MRD negativity.

Efficacy – OS

- At the time of analysis, OS data were not yet mature.
- In patients aged ≥75 years, 8/32 (25.0%) in the Isa-Pd arm and 15/29 (51.7%) in the Pd arm had died.
 - Median OS was not reached in the Isa-Pd arm and was 10.3 months in the Pd arm with a CI that does not cross 1 (HR 0.40; 95% CI 0.17–0.96).

Figure 2. PFS in the Isa-Pd and Pd arms in patients ≥75 years

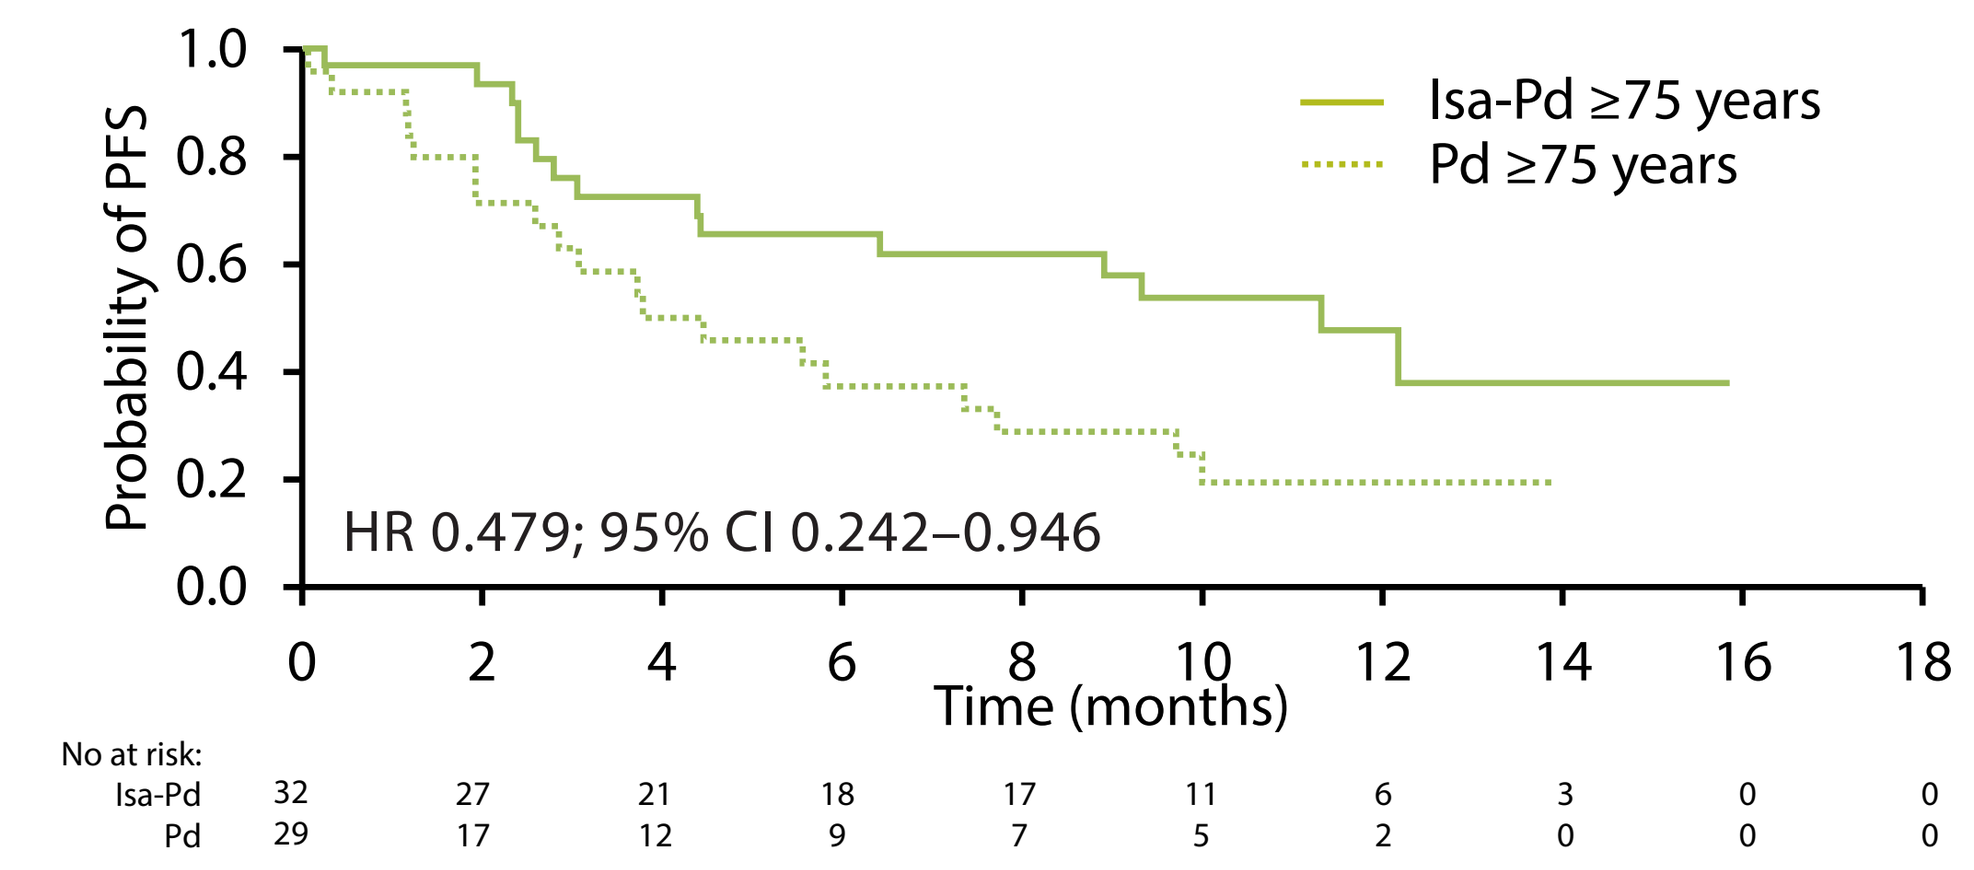

Probability of PFS
Time (months)
Isa-Pd ≥75 years
Pd ≥75 years
HR 0.479; 95% CI 0.242–0.946
No at risk:
Isa-Pd 32 27 21 18 17 11 6 3 0 0
Pd 29 17 12 9 7 5 2 0 0 0

CI, confidence interval; d, dexamethasone; HR, hazard ratio; Isa, isatuximab; P, pomalidomide; PFS, progression-free survival.

Figure 3. PFS in the Isa-Pd and Pd arms in patients 65–74 years

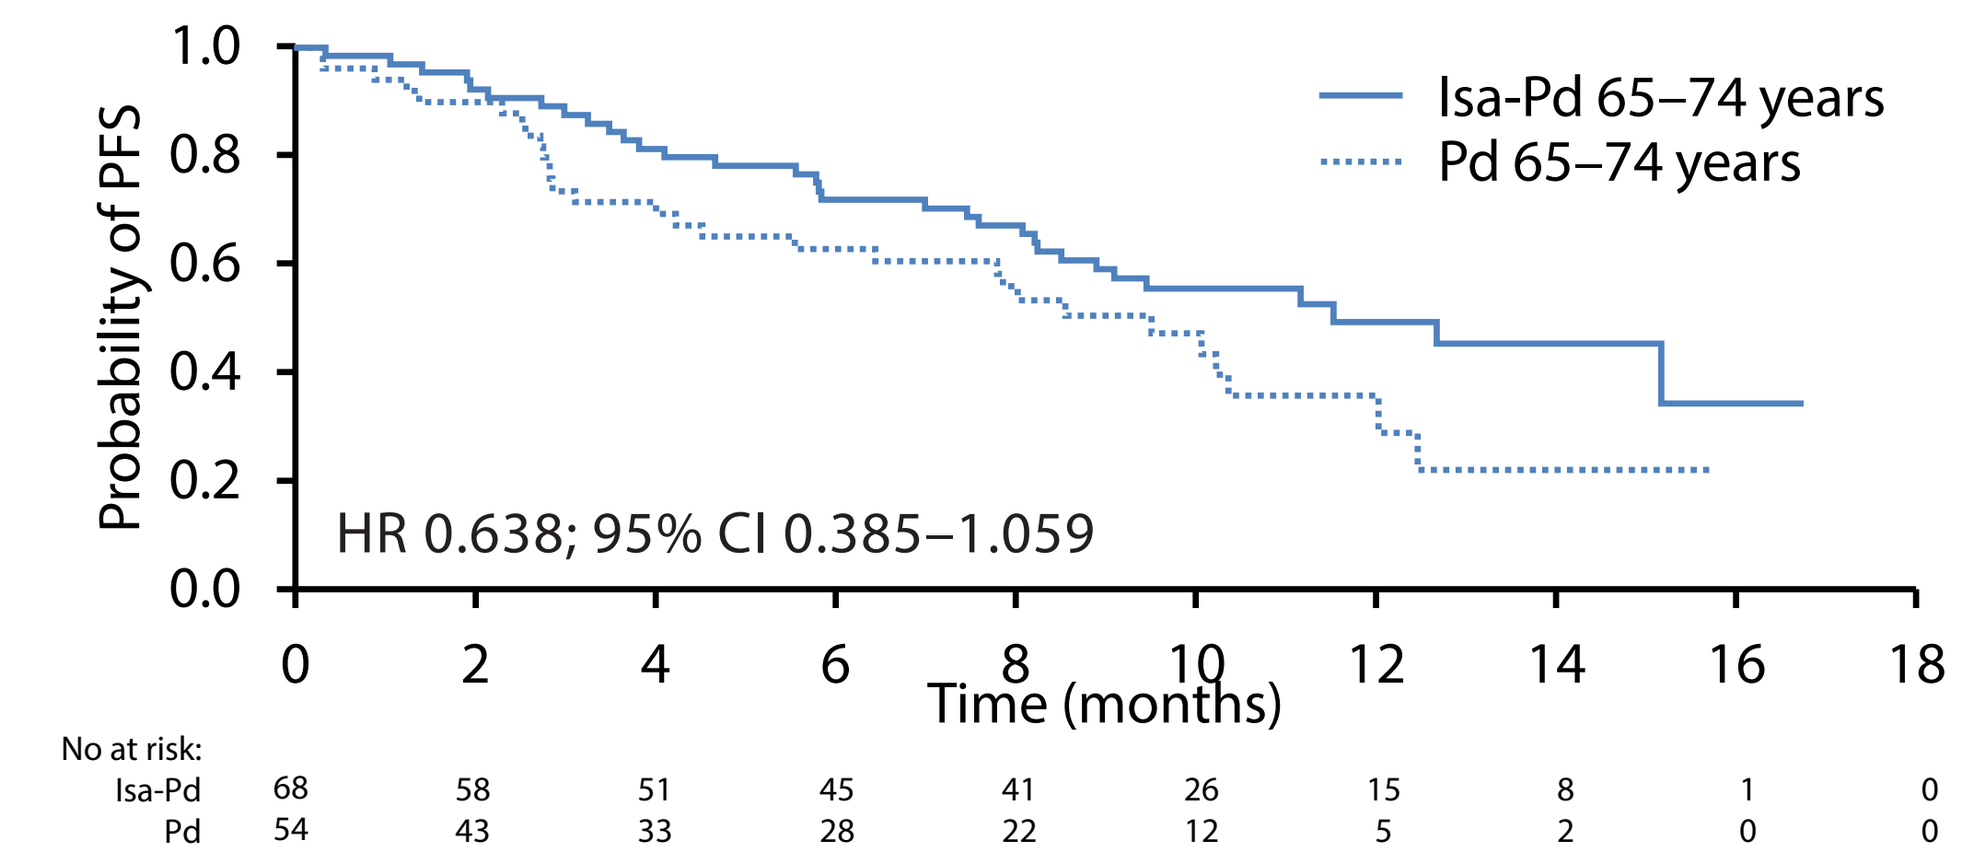

Probability of PFS
Time (months)
Isa-Pd 65–74 years
Pd 65–74 years
HR 0.638; 95% CI 0.385–1.059
No at risk:
Isa-Pd 68 58 51 45 41 26 15 8 1 0
Pd 54 43 33 28 22 12 5 2 0 0

CI, confidence interval; d, dexamethasone; HR, hazard ratio; Isa, isatuximab; P, pomalidomide; PFS, progression-free survival.

Figure 4. PFS in the Isa-Pd and Pd arms in patients <65 years

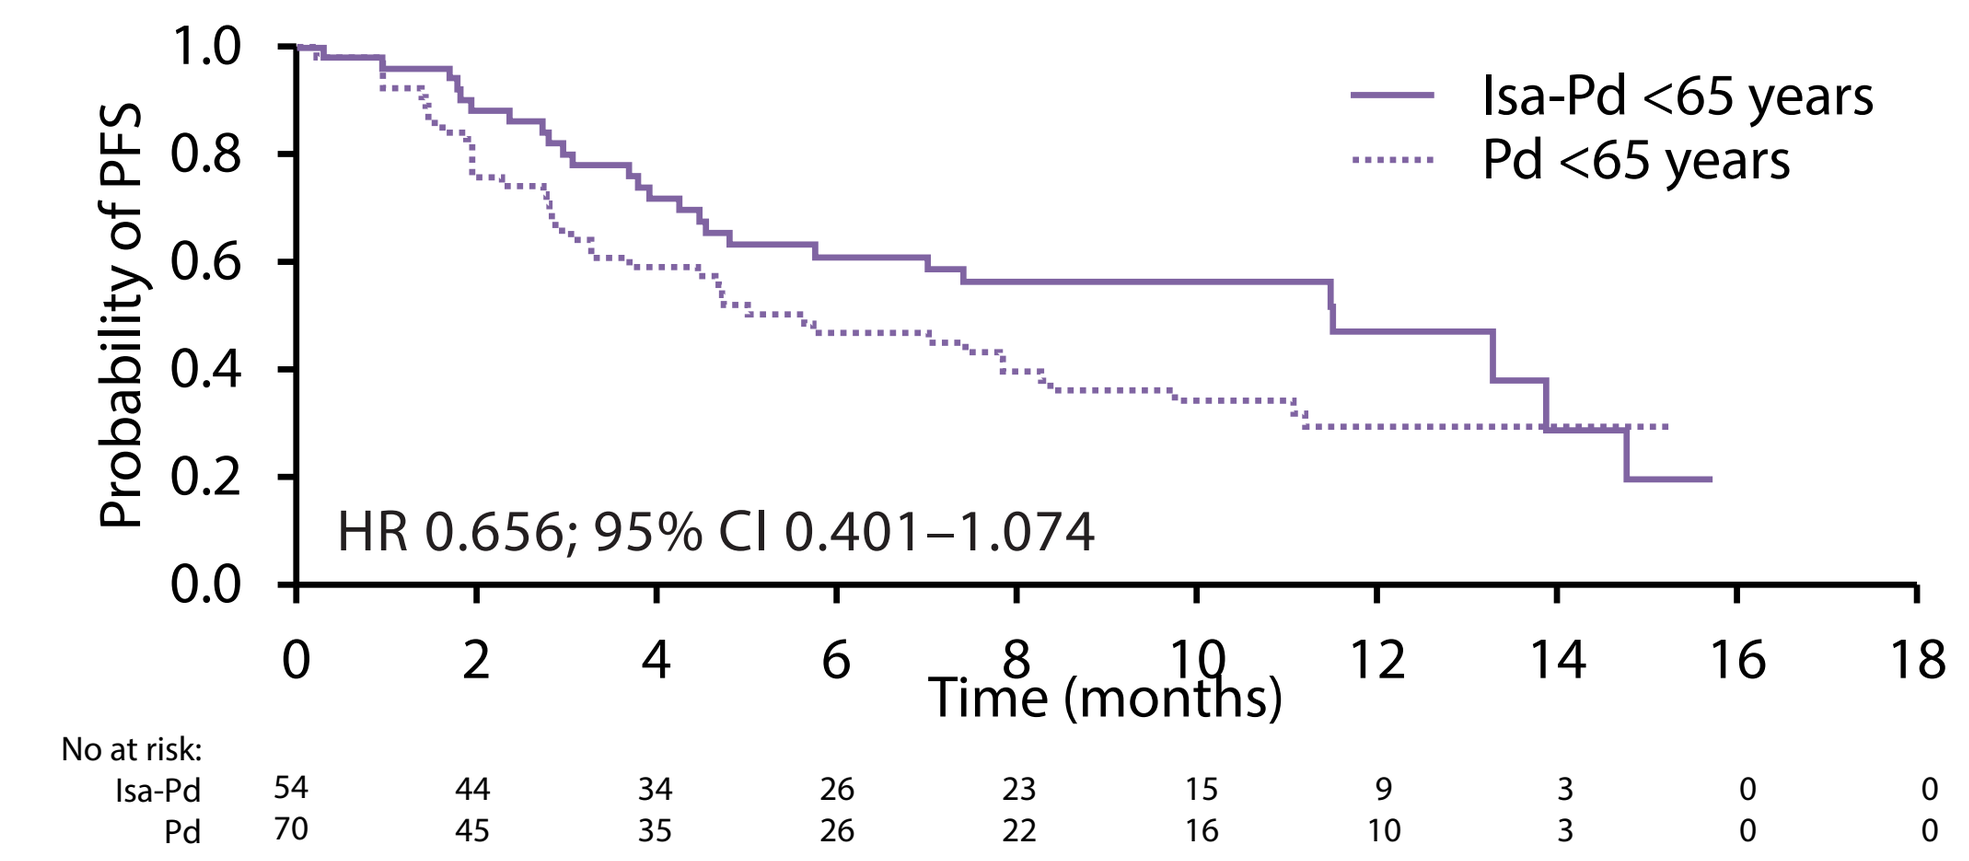

Probability of PFS
Time (months)
Isa-Pd <65 years
Pd <65 years
HR 0.656; 95% CI 0.401–1.074
No at risk:
Isa-Pd 54 44 34 26 23 15 9 3 0 0
Pd 70 45 35 26 22 16 10 3 0 0

CI, confidence interval; d, dexamethasone; HR, hazard ratio; Isa, isatuximab; P, pomalidomide; PFS, progression-free survival.

Table 1. Most common TEAEs (≥15% all grades or ≥10% grade ≥3) by patient age group

TEAE, n (%)	Age ≥75 years				Age 65–74 years				Age <65 years			
	Isa-Pd (n=32)		Pd (n=28)		Isa-Pd (n=66)		Pd (n=53)		Isa-Pd (n=54)		Pd (n=68)	
	All grades	Grade ≥3	All grades	Grade ≥3	All grades	Grade ≥3	All grades	Grade ≥3	All grades	Grade ≥3	All grades	Grade ≥3
Neutropenia	17 (53.1)	16 (50.0)	13 (46.4)	13 (46.4)	30 (45.5)	30 (45.5)	18 (34.0)	17 (32.1)	24 (44.4)	24 (44.4)	19 (27.9)	18 (26.5)
Pneumonia	4 (12.5)	4 (12.5)	2 (7.1)	2 (7.1)	18 (27.3)	14 (21.2)	7 (13.2)	7 (13.2)	9 (16.7)	7 (13.0)	17 (25.0)	14 (20.6)
Upper respiratory tract infection	10 (31.3)	1 (3.1)	1 (3.6)	0	22 (33.3)	1 (1.5)	8 (15.1)	1 (1.9)	11 (20.4)	3 (5.6)	17 (25.0)	0
Diarrhea	12 (37.5)	1 (3.1)	7 (25.0)	0	14 (21.2)	1 (1.5)	10 (18.9)	1 (1.9)	13 (24.1)	1 (1.9)	12 (17.6)	0
Fatigue	5 (15.6)	2 (6.3)	7 (25.0)	0	16 (24.2)	3 (4.5)	10 (18.9)	0	5 (9.3)	1 (1.9)	15 (22.1)	0
Infusion related reaction	9 (28.1)	1 (3.1)	0	0	24 (36.4)	2 (3.0)	1 (1.9)	0	23 (42.6)	1 (1.9)	1 (1.5)	0
Asthenia	5 (15.6)	1 (3.1)	7 (25.0)	2 (7.1)	9 (13.6)	3 (4.5)	9 (17.0)	0	9 (16.7)	1 (1.9)	11 (16.2)	2 (2.9)
Constipation	4 (12.5)	0	7 (25.0)	0	11 (16.7)	0	7 (13.2)	0	9 (16.7)	0	12 (17.6)	0
Bronchitis	8 (25.0)	1 (3.1)	1 (3.6)	1 (3.6)	17 (25.8)	4 (6.1)	5 (9.4)	0	11 (20.4)	0	7 (10.3)	0
Back pain	6 (18.8)	0	6 (21.4)	1 (3.6)	10 (15.2)	1 (1.5)	4 (7.5)	0	9 (16.7)	2 (3.7)	12 (17.6)	1 (1.5)
Pyrexia	7 (21.9)	1 (3.1)	7 (25.0)	1 (3.6)	9 (13.6)	1 (1.5)	5 (9.4)	1 (1.9)	6 (11.1)	0	9 (13.2)	0
Dyspnea	7 (21.9)	0	5 (17.9)	0	7 (10.6)	3 (4.5)	6 (11.3)	1 (1.9)	9 (16.7)	3 (5.6)	4 (5.9)	1 (1.5)
Thrombocytopenia	6 (18.8)	5 (15.6)	3 (10.7)	3 (10.7)	9 (13.6)	9 (13.6)	7 (13.2)	7 (13.2)	4 (7.4)	4 (7.4)	8 (11.8)	8 (11.8)
Urinary tract infection	2 (6.3)	1 (3.1)	8 (28.6)	1 (3.6)	8 (12.1)	3 (4.5)	2 (3.8)	0	5 (9.3)	3 (5.6)	4 (5.9)	1 (1.5)
Febrile neutropenia	4 (12.5)	4 (12.5)	0	0	8 (12.1)	8 (12.1)	1 (1.9)	1 (1.9)	6 (11.1)	6 (11.1)	2 (2.9)	2 (2.9)
Acute kidney injury	5 (15.6)	2 (6.3)	3 (10.7)	2 (7.1)	1 (1.5)	1 (1.5)	1 (1.9)	1 (1.9)	1 (1.9)	1 (1.9)	4 (5.9)	3 (4.4)

Per eCRF completion guidelines, laboratory abnormalities are reported as adverse event only if leading to an investigational medicinal product administration change (dose delayed, dose reduced, drug interrupted and/or drug withdrawn), and/or fulfilling a seriousness criterion
d, dexamethasone; Isa, isatuximab; P, pomalidomide; TEAE, treatment-emergent adverse event.

Figure 5. Response in the Isa-Pd and Pd arms by patient age group

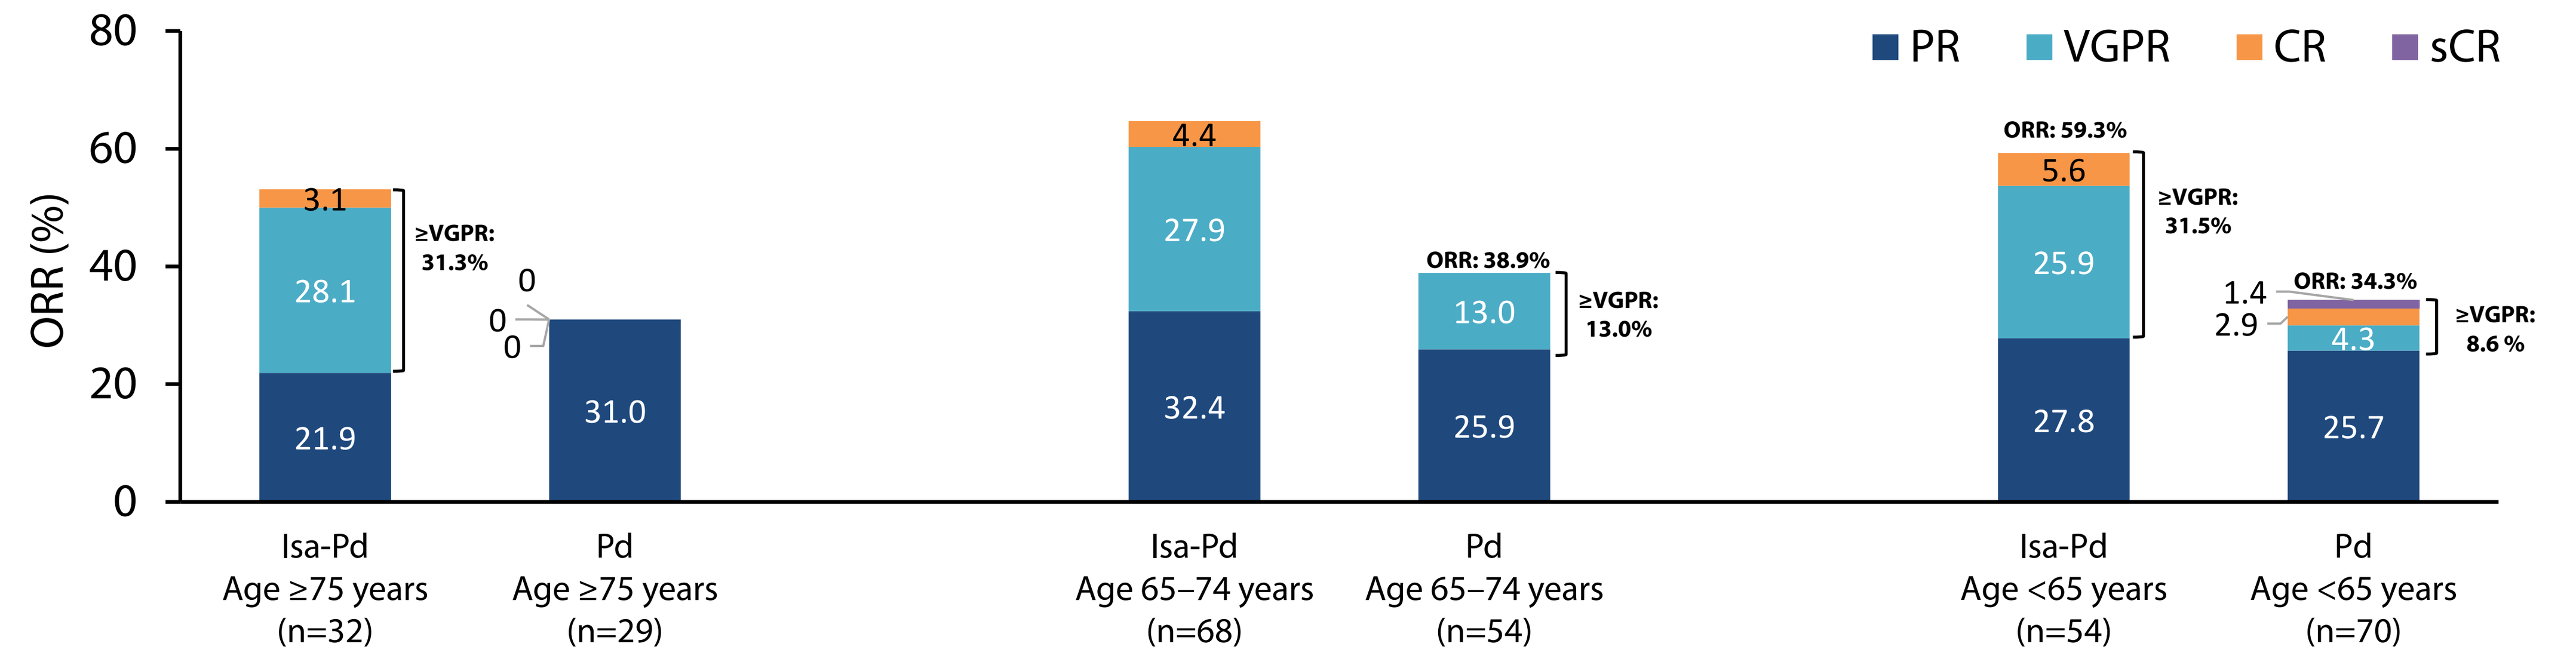

PR
VGPR
CR
sCR
ORR (%)
3.1
28.1
21.9
≥VGPR: 31.3%
0
0
0
31.0
4.4
27.9
32.4
ORR: 38.9%
13.0
25.9
≥VGPR: 13.0%
ORR: 59.3%
5.6
25.9
27.8
≥VGPR: 31.5%
ORR: 34.3%
1.4
2.9
4.3
25.7
≥VGPR: 8.6 %
Isa-Pd Age ≥75 years (n=32)
Pd Age ≥75 years (n=29)
Isa-Pd Age 65–74 years (n=68)
Pd Age 65–74 years (n=54)
Isa-Pd Age <65 years (n=54)
Pd Age <65 years (n=70)

CR, complete response; d, dexamethasone; Isa, isatuximab; ORR, overall response rate; P, pomalidomide; PR, partial response; sCR, stringent complete response; VGPR, very good partial response.

Efficacy – OS (cont'd)

- For patients aged 65–74 years, median OS was not reached in the Isa-Pd arm and was 14.5 months in the Pd arm (HR 0.75; 95% CI 0.38–1.45).
- Median OS was not reached for either treatment arm in the group of patients aged <65 years. The HR for Isa-Pd vs Pd was 0.85 (95% CI 0.46–1.59).
- One-year OS rates for the different age groups were similar for Isa-Pd; 73.5%, 74.7% and 67.7% for ≥75, 65–74 and <65 years, respectively. One-year OS rates for Pd were 47.2%, 72.9% and 63.0%, respectively.

Safety

- The number of patients with any TEAE was similar with Isa-Pd vs Pd (98.1% vs 97.1% for patients aged <65 years; 100% vs 98.1% for patients aged 65–74 years). All patients aged ≥75 years in both treatment groups experienced TEAEs.
- In the Isa-Pd arm, the most common any grade non-hematologic TEAE was infusion related reaction, regardless of age group (**Table 1**).
 - There was a trend towards fewer infusion reactions in patients ≥75 years (28.1%) compared with patients 65–74 years (36.4%) or <65 years (42.6%)
- In the Isa-Pd arm, TEAEs with the greatest difference in incidence for patients aged ≥75 years vs <65 years were infusion related reaction (28.1% vs 42.6%) and acute kidney injury (15.6% vs 1.9%) (**Table 1**).
- In both treatment groups, Grade ≥3 TEAEs were more frequent in patients aged ≥75 years than those aged <65 years (93.8% vs 85.2% with Isa-Pd; 75.0% vs 64.7% with Pd).
- The most common grade ≥3 non-hematologic TEAE was pneumonia, regardless of patient age or treatment group (**Table 1**).
- Grade 3–4 neutropenia or thrombocytopenia, based on laboratory results, were more common with Isa-Pd than Pd, regardless of patient age group (**Table 2**). Grade 4 thrombocytopenia was more common with Pd in patients aged <65 years (**Table 2**).
- No patient experienced hemorrhage after Grade 4 thrombocytopenia.

Table 2. Abnormal hematologic parameters (grade 3–4) during treatment by patient age group

Hematologic abnormality, n (%)	Age ≥75 years		Age 65–74 years		Age <65 years	
	Isa-Pd (n=32)	Pd (n=28)	Isa-Pd (n=66)	Pd (n=53)	Isa-Pd (n=54)	Pd (n=68)
Neutropenia	28 (87.5)	18 (64.3)	53 (80.3)	38 (71.7)	48 (88.9)	47 (69.1)
Thrombocytopenia	11 (34.4)	8 (28.6)	20 (30.3)	13 (24.5)	16 (29.6)	15 (22.1)
Grade 4	6 (18.8)	3 (10.7)	13 (19.7)	9 (17.0)	6 (11.1)	10 (14.7)

d, dexamethasone; Isa, isatuximab; P, pomalidomide.

- In both treatment arms TEAEs leading to treatment discontinuation were more common in patients aged ≥75 years vs <65 years (15.6% vs 7.4% with Isa-Pd; 14.3% vs 10.3% with Pd), with no difference between arms.
 - Among patients aged 65–74 years TEAEs leading to treatment discontinuation were 3.0% vs 15.1%, respectively.
- In both treatment arms there was a greater incidence of serious TEAEs (SAEs) in patients aged ≥75 years vs <65 years (68.8% vs 57.4% with Isa-Pd; 57.1% vs 47.1% with Pd).
- The incidence of TEAEs with a fatal outcome was lower in patients aged ≥75 years vs <65 years in the Isa-Pd arm (6.3% vs 11.1%), but the opposite trend was seen in the Pd arm (14.3% vs 5.9%).

CONCLUSIONS

- PFS, ORR, ≥VGPR, and OS were all improved in elderly patients with the addition of Isa to Pd, with similar PFS and 1-year OS observed in elderly and younger patients.
- The benefits of Isa-Pd vs Pd in elderly patients were consistent with those observed in the overall ICARIA-MM study population.
- The incidence of SAEs and discontinuations due to TEAEs was higher in patients aged ≥75 years compared with younger patients with both Isa-Pd and Pd, but there was no increase in fatal adverse events in the Isa-Pd arm.

ACKNOWLEDGMENTS
The ICARIA-MM study was sponsored by Sanofi.
The authors thank the participating patients and their families, and the study centers and investigators, for their contributions to the study.
Medical writing support was provided by Beatrice Tyrrell, DPhil, of Aspire Scientific (Bollington, UK), and funded by Sanofi (Cambridge, MA, USA).

DISCLOSURES
FHS: Research funding – Celgene; honoraria – Amgen, Celgene, Takeda, Janssen, Novartis, and SkyliteDX; membership on an entity's Board of Directors or advisory committees – Amgen, Celgene, Takeda, Janssen, Oncopeptides, and MSD. **PGR**: Research funding – Bristol-Myers Squibb, Celgene, Oncopeptides, and Takeda; membership on an entity's Board of Directors or advisory committees – Amgen, Celgene, Janssen, Karyopharm, Oncopeptides, Sanofi, and Takeda. **TF**: Membership on an entity's Board of Directors or advisory committees – Amgen, Celgene, Janssen, Karyopharm, Oncopeptides, Roche, Sanofi, and Takeda; Speakers Bureau – Celgene, Janssen, and Takeda. **AS**: Research funding – AbbVie, Amgen, Celgene, Janssen Oncology, and Takeda; honoraria – Specialised Therapeutics Australia; Speakers Bureau – Celgene; consulting/advisory role – AbbVie, Amgen, Celgene, Haemalogix, Janssen Oncology, Sanofi, Secura Bio, Servier, and Takeda; consultancy – Specialised Therapeutics Australia. **KS**: research funding – AbbVie, Alexion-pharma, Bristol-Myers Squibb, Celgene, Daiichi Sankyo, GSK, Janssen, MSD, Novartis, Ono, Sanofi, and Takeda; honoraria – Bristol-Myers Squibb, Celgene, Ono, and Takeda. **SBr**: honoraria – Amgen, Bristol-Myers Squibb, Celgene Corporation, and Janssen; membership on an entity's Board of Directors or advisory committees – Amgen, Celgene Corporation, Janssen, and Karyopharm; consultancy – Janssen and Takeda. **SLG**, **FC**, **HvdV**, and **SBe** are employed by Sanofi. **MA**, **AA**, **AJ**, **LF**, and **CKM** have no relevant financial relationships to disclose.

REFERENCES
1. National Cancer Institute. https://seer.cancer.gov/statfacts/html/mulmy.html.
2. Willan J, et al. Clin Interv Aging 2016;11:423–35.
3. Bringhen S, et al. Haematologica 2013;98:980–7.
4. Deckert J, et al. Clin Cancer Res 2014;20:4574–83.
5. Jiang H, et al. Leukemia 2016;30:399–408.
6. Richardson P, et al. Future Oncol 2018;14:1035–47.
7. Attal M, et al. Lancet 2019 – manuscript in press.

Scan for this poster